UNI-494 Lowers Urine β2-Microglobulin in Rat Ischemia/Reperfusion Model

Guru Reddy, PhD[1], Pramod Gupta, PhD[1], Atul Khare, PhD[1], Shalabh Gupta, MD[1]

1 Unicycive Therapeutics, Inc.

UNICYCIVE THERAPEUTICS INC.

Background

- Mitochondrial dysfunction in renal cells plays a critical role in the pathophysiology of acute kidney injury (AKI) and chronic kidney disease (CKD)[1]
- The proximal tubule is the primary sensor and effector in CKD progression and AKI,[2] and measurement of urine β2-microglobulin (β2M) is a sensitive assay for proximal tubule injury[3]
- Nicorandil, a selective mitochondrial ATP-sensitive potassium channel activator,[4] **(Figure 1)** may be a promising AKI treatment,[5] but its clinical use is limited by serious gastrointestinal side effects and rapid absorption and elimination[6,7]
- UNI-494, a novel nicorandil prodrug designed to improve its pharmacologic properties, may increase the exposure to nicorandil and improve the safety profile of nicorandil

Figure 1: Mechanism of Nicorandil Blocks MPTP Pores by Binding to K_{ATP} Channel

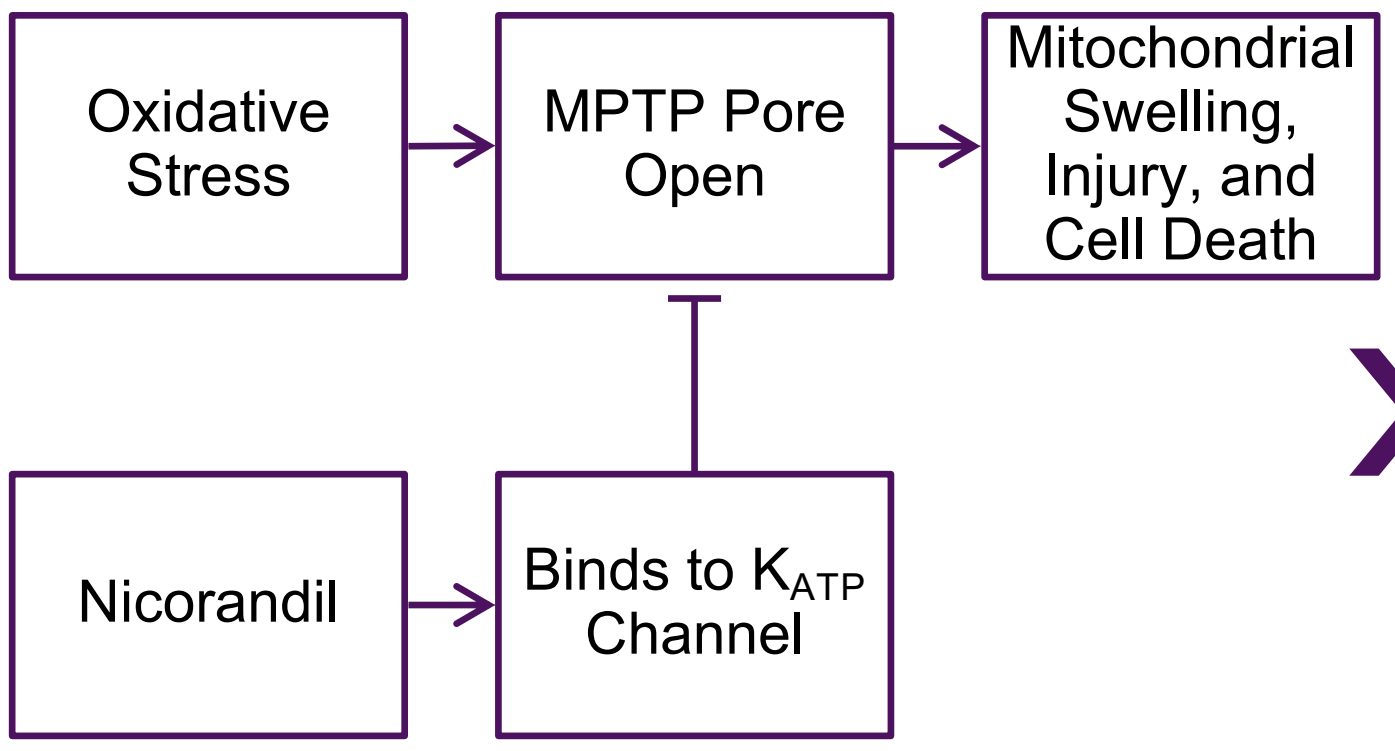
Oxidative Stress
MPTP Pore Open
Mitochondrial Swelling, Injury, and Cell Death
Nicorandil
Binds to K_{ATP} Channel

KEY OBSERVATIONS

- Indicated for the treatment of angina
- Approved ex-U.S. for ~40 years
- Dual mechanism in angina – NO donor and ATP-sensitive K^+ channel (K_{ATP}) agonist
- Widely published evidence that nicorandil exhibits broad mitochondrial protection
- Primary mechanism likely via activation of mitochondrial K_{ATP} channel

Objective

We present efficacy data from a study of UNI-494 in a rat ischemia-reperfusion (I/R) model

Methods and Materials

- 49 male Sprague-Dawley rats were randomly assigned to 4 groups to evaluate the *in vivo* efficacy of UNI-494 in a bilateral renal I/R model
- Group 1 was the Sham group (n=10) and groups 2-4 (n=13 each, 10+3 back-ups) were established as the I/R models (45 minutes of bilateral occlusion)
- Group 1 received no treatment, Group 2 received vehicle, Group 3 received UNI-494 10 mg/kg, and Group 4 received UNI-494 20 mg/kg
- Treatments were administered as a single dose on Day 0, 1 hour prior to modeling
- Body weights were measured on Days -1, 0, and 1
- Urine samples were collected within 24 hours after the surgery using metabolic cages
- T-tests were used to evaluate statistical differences between groups ($p<0.05$ was considered significant)

Results

- β2M urine levels were significantly lower for the UNI-494 20 mg/kg dose group compared to the vehicle group
- β2M total urine content in both UNI-494 dose groups (10 and 20 mg/kg) was significantly lower compared to the vehicle group **(Figure 2)**
- β2M urine levels and content were lower for the 20 mg/kg UNI-494 dose group than for the 10 mg/kg dose group **(Figure 2)**

Results (Continued)

- No physical or behavioral abnormalities, significant changes in body weight **(Figure 3)**, or significant differences in histological scores were observed between UNI-494 groups and vehicle

Figure 2: Mean (±SEM) Comparison of Urine β2M Content Between Groups

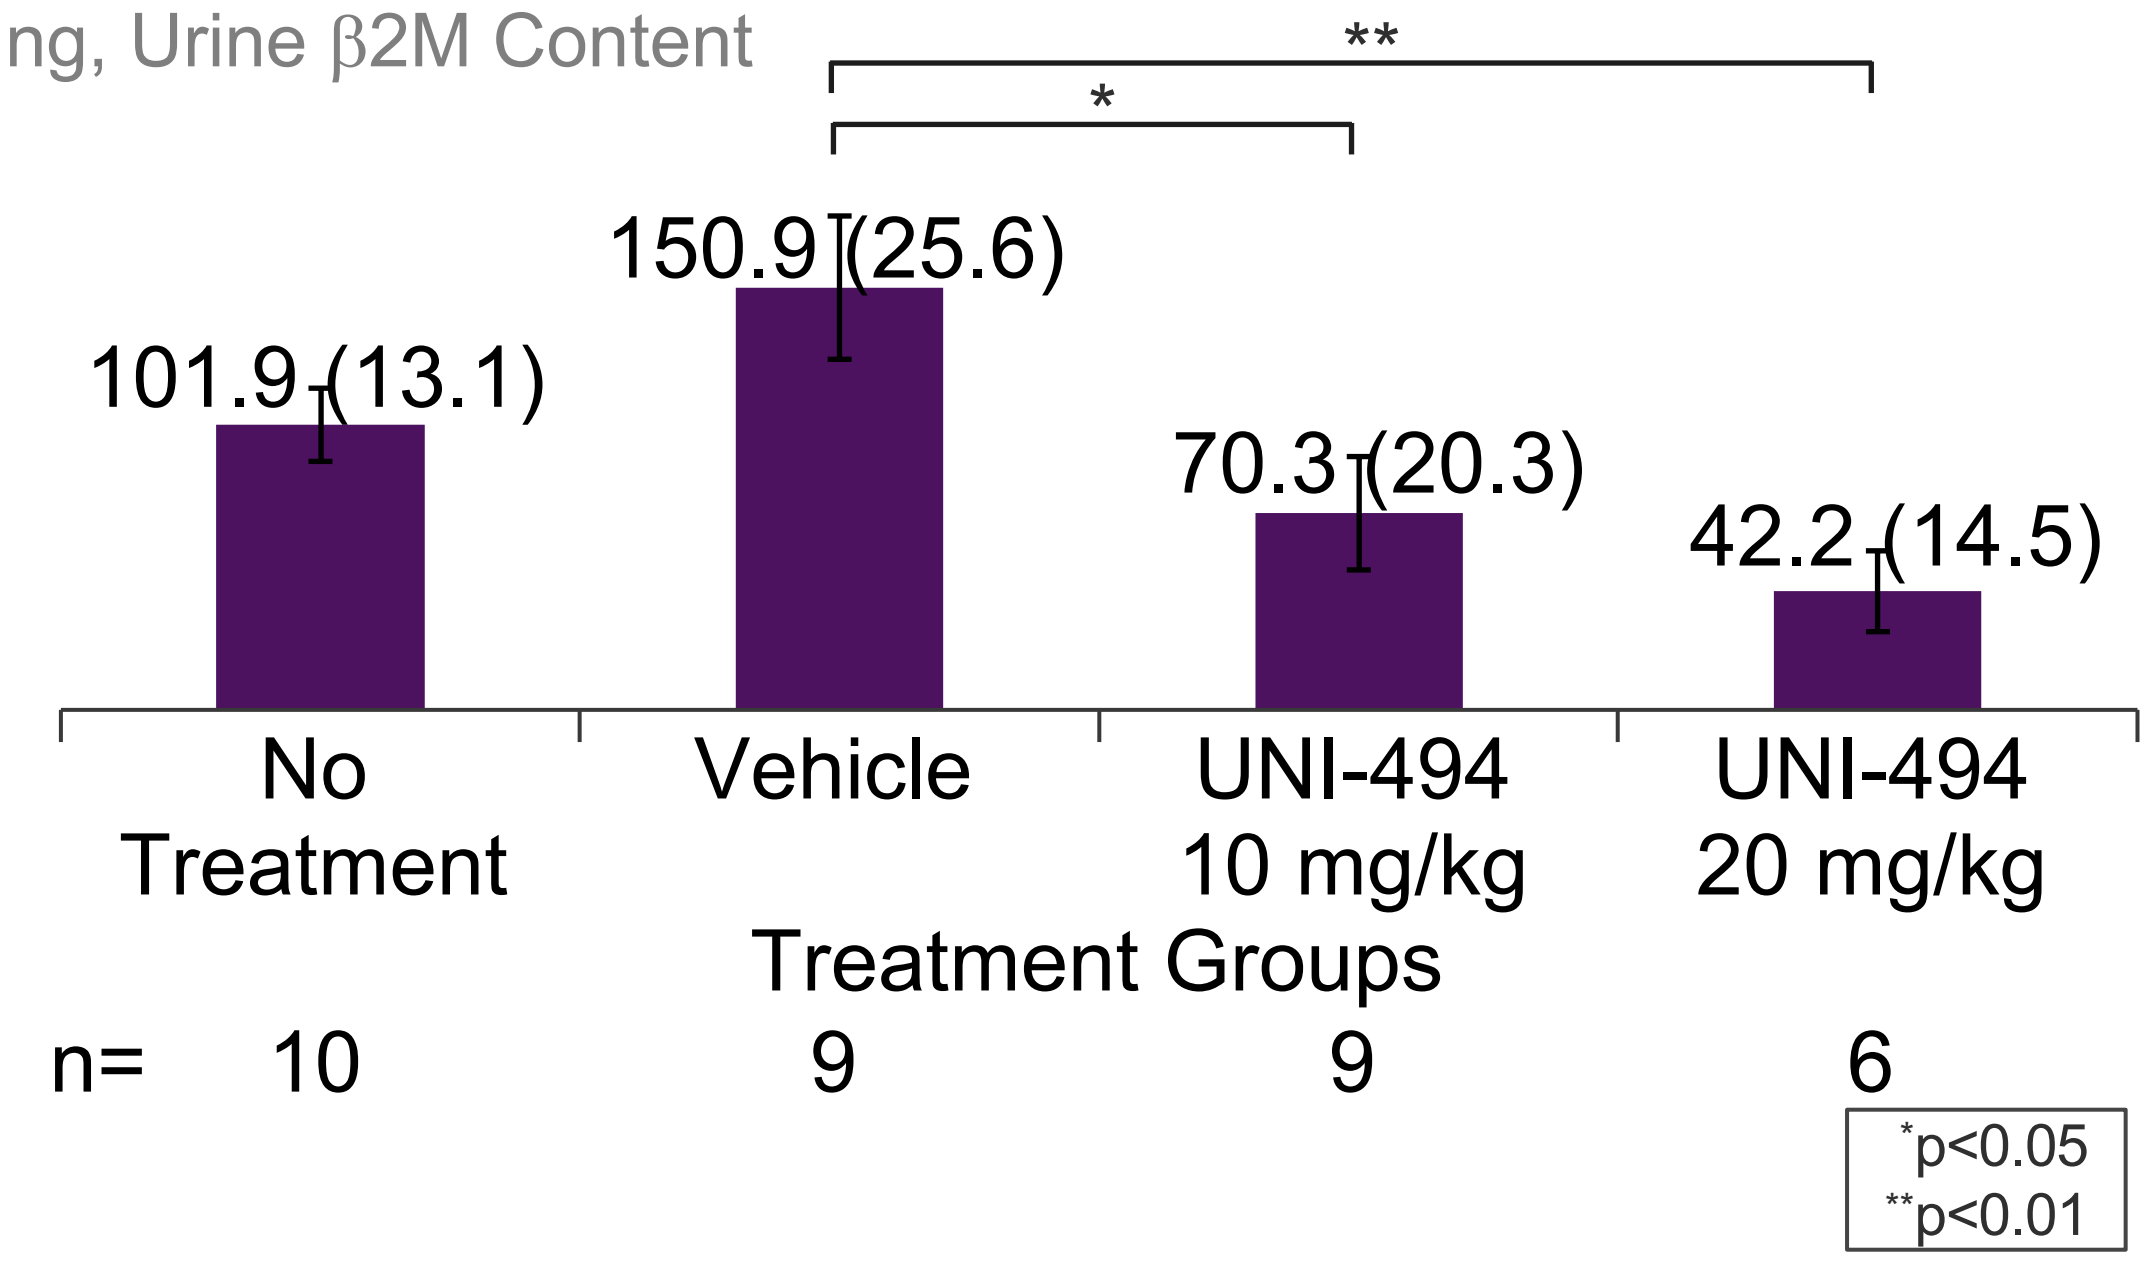
ng, Urine β2M Content
**
*
101.9 (13.1)
150.9 (25.6)
70.3 (20.3)
42.2 (14.5)
No Treatment
Vehicle
UNI-494 10 mg/kg
UNI-494 20 mg/kg
Treatment Groups
n= 10 9 9 6
*p<0.05
**p<0.01

Figure 2: Mean (±SEM) Comparison of Body Weight Between Groups[1]

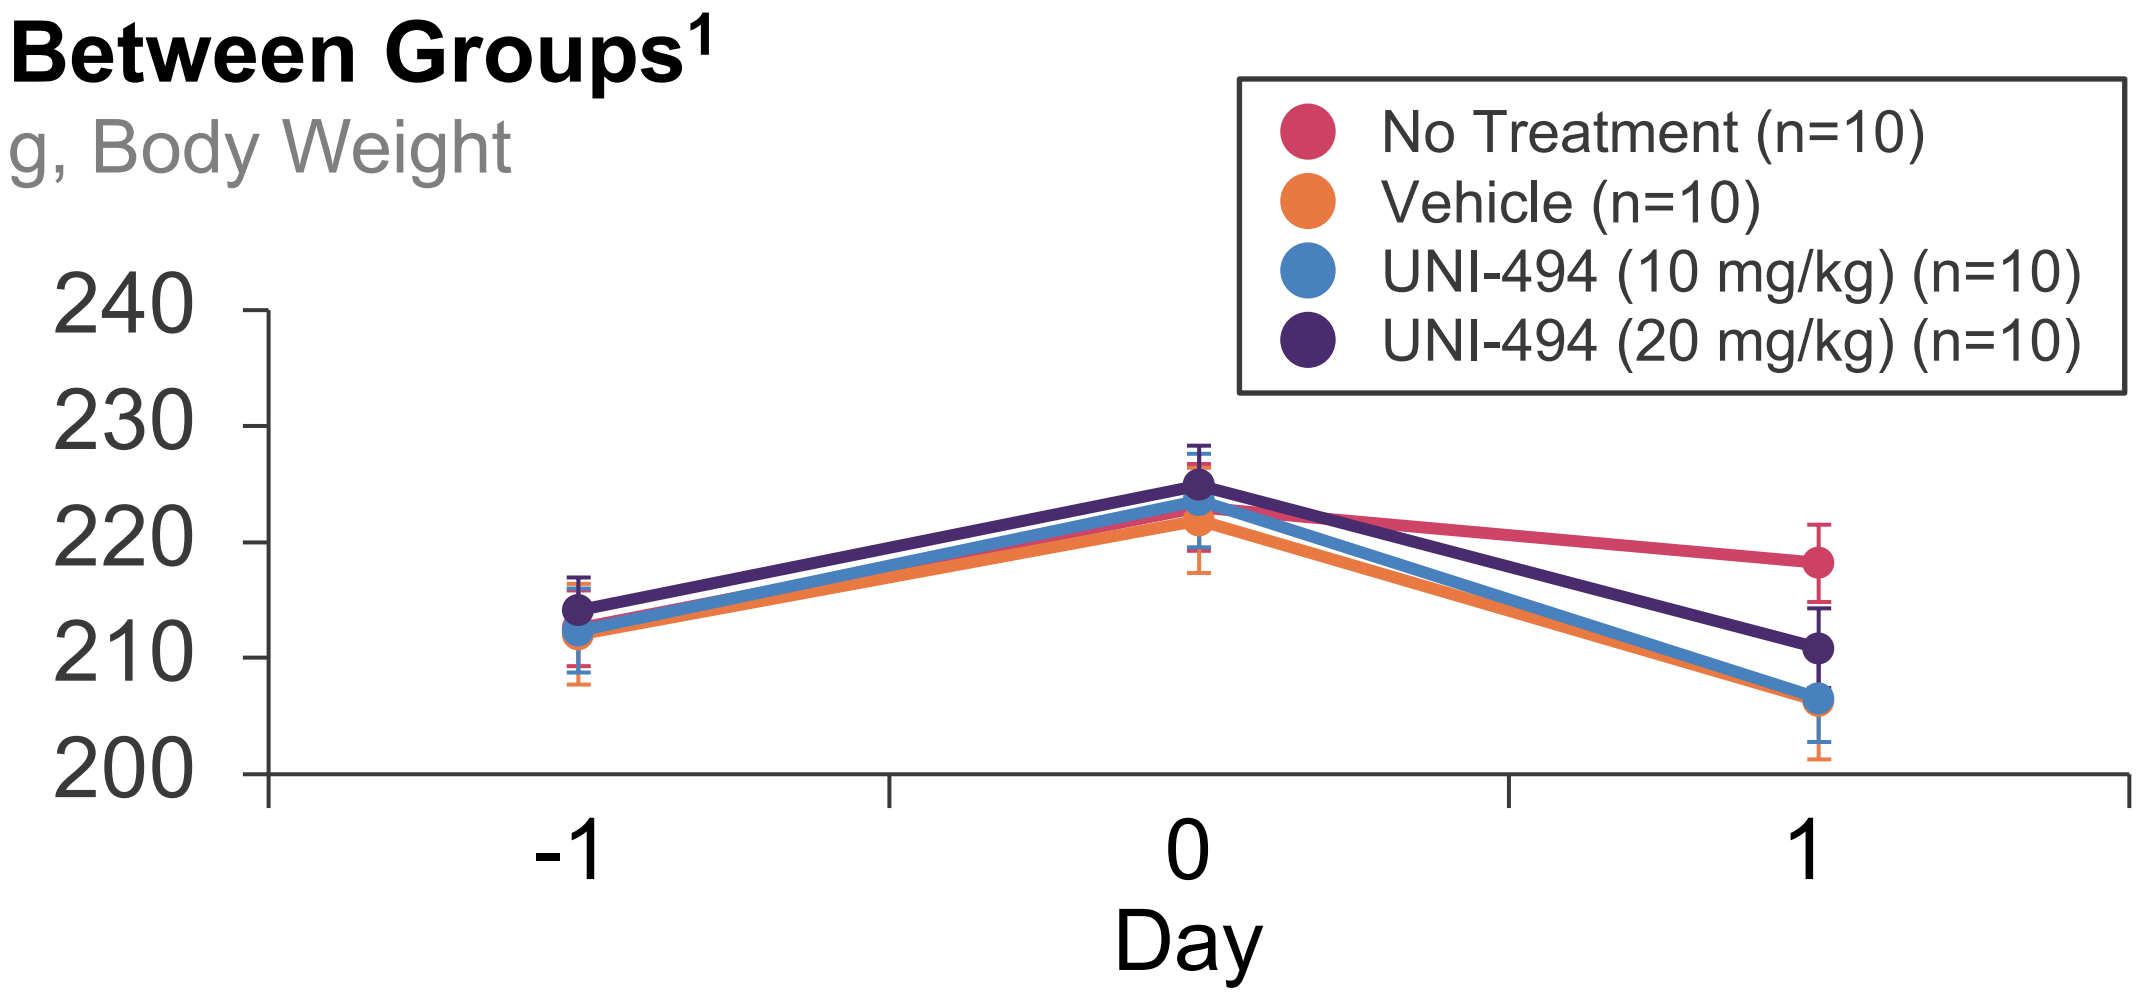
g, Body Weight
No Treatment (n=10)
Vehicle (n=10)
UNI-494 (10 mg/kg) (n=10)
UNI-494 (20 mg/kg) (n=10)
240
230
220
210
200
-1
0
1
Day

1 No significant difference using one-way ANOVA

Conclusions

- The lower level of urine β2M in the higher dose group and lower total urine β2M content in both UNI-494 dose groups, compared to vehicle, indicate that UNI-494 may have a renoprotective effect
- The lower urine β2M content in the higher dose group (20 mg/kg) compared to the lower dose group (10 mg/kg) indicate a dose-response trend

Implications

The mechanism of this potential renoprotective effect of UNI-494 should be further investigated

References:
1. Zhang X et al., *Int J Mol Sci.* 2021.
2. Chevalier RL., *Am J Physiol Renal Physiol*. 2016.
3. Zeng X et al., *J Biomark.* 2014.
4. Kuno A et al., *Basic Res Cardiol.* 2007.
5. Rabea M et al., *QJM: An International Journal of Medicine*. 2021.
6. Frydman AM et al., *American Journal of Cardiology*. 1989.
7. SANOFI. IKOREL 10mg and 20mg Tablets (nicorandil) Package Leaflet. 2021.

Acknowledgements:
Writing support was provided by Xelay Acumen Group, Inc., and funded by Unicycive Therapeutics, Inc.

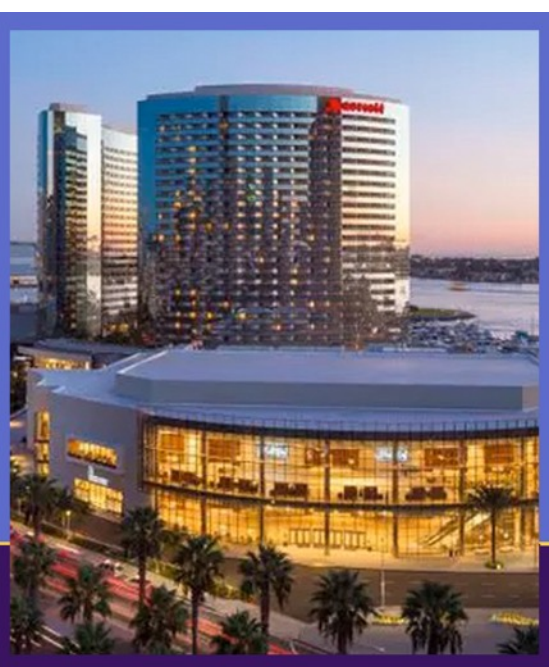

THE 28TH INTERNATIONAL CONFERENCE ON
ADVANCES IN CRITICAL CARE NEPHROLOGY
AKI & CRRT 2023
MARCH 29 - APRIL 1 SAN DIEGO, CALIFORNIA